EFFICACY AND TOLERABILITY OF MERATRIM FOR WEIGHT MANAGEMENT: A RANDOMIZED, DOUBLE-BLIND, PLACEBO-CONTROLLED STUDY IN HEALTHY OVERWEIGHT SUBJECTS.

Zainulabedin M. Saiyed[1], Golakoti Trimurtulu[2], Alluri V. Krishnaraju[2] and James P. Lugo[1]

[1]InterHealth Nutraceuticals, Benicia, CA; [2]Laila Nutraceuticals, Vijayawada, India

SPECIFIC AIM

Meratrim® is a blend of two plant extracts, Sphaeranthus indicus flower heads and Garcinia mangostana fruit rind. Previous preclinical and clinical studies have demonstrated that Meratrim is safe and effective for weight management in obese individuals. The objective of this study was to assess the efficacy and tolerability of Meratrim in managing body weight in healthy overweight subjects with a body mass index (BMI) of 27-32 kg/m^2. The primary endpoint was defined as the change in body weight from baseline to week 16 for the Meratrim group versus placebo.

MATERIALS & METHODS

MATERIAL

- Meratrim® and sensory identical placebo capsules were provided by Laila Nutraceuticals (Vijayawada, India).

TRIAL LOCATION

- Study was run & managed by an independent clinical research organization D2L (Bangalore, India).
- The trial was conducted at a single site, Srinivasa Clinic & Diabetic Care Center, Bangalore, India.
- The study protocol was approved by Bangalore Ethics Committee, and listed on the clinical trial registry of India (CTRI/2014/07/004727).

TRIAL DESIGN

- Randomized, double-blind, placebo-controlled clinical study.
- Subjects (age 21-50 years) with BMI between 27 and 32 kg/m^2 were recruited in the study.
- Sixty subjects were randomly divided into two groups receiving a daily dose of Meratrim (400 mg twice daily) or placebo over a 16-week period.
- Fifty seven subjects completed the trial: 29, Meratrim; 28, placebo.
- Study assessments were done at 0 (baseline), 2, 4, 8, 12 and 16 weeks.
- Subjects were asked to consume 2,000 kcal/day diet (17% protein, 25% fat and 58% carbohydrate).
- Subjects were instructed to perform walking exercise for 30 minutes 5 days per week.
- All subjects completed an appetite and mood questionnaire (VAS & POMS-SF) at baseline, weeks 8 and 16.
- Efficacy and safety measurements included body weight, BMI, waist and hip size, body composition, blood lipid profile, vital signs, blood chemistry, hematology and adverse event recording.

STATISTICAL ANALYSIS

- Intent-to-treat and per protocol analysis was performed for all endpoints using analysis of covariance (ANCOVA) with the baseline as a covariate.
- Data is presented as mean ± SE. $p \leq 0.05$ was considered statistically significant.
- Statistical analyses were performed using SAS version 9.2 (Cary, NC).

RESULTS

- At study conclusion, the Meratrim supplemented group demonstrated a statistically significant reduction in body weight versus placebo (Figure 1).
- Waist and hip size was significantly reduced in the Meratrim cohort compared to placebo at 16 weeks (Figure 2).
- A statistically significant improvement in blood lipid profile was seen in the Meratrim group compared to placebo (Table 1).
- No adverse events related to supplementation were noted during the study.

Figure 1: Reduction in body weight

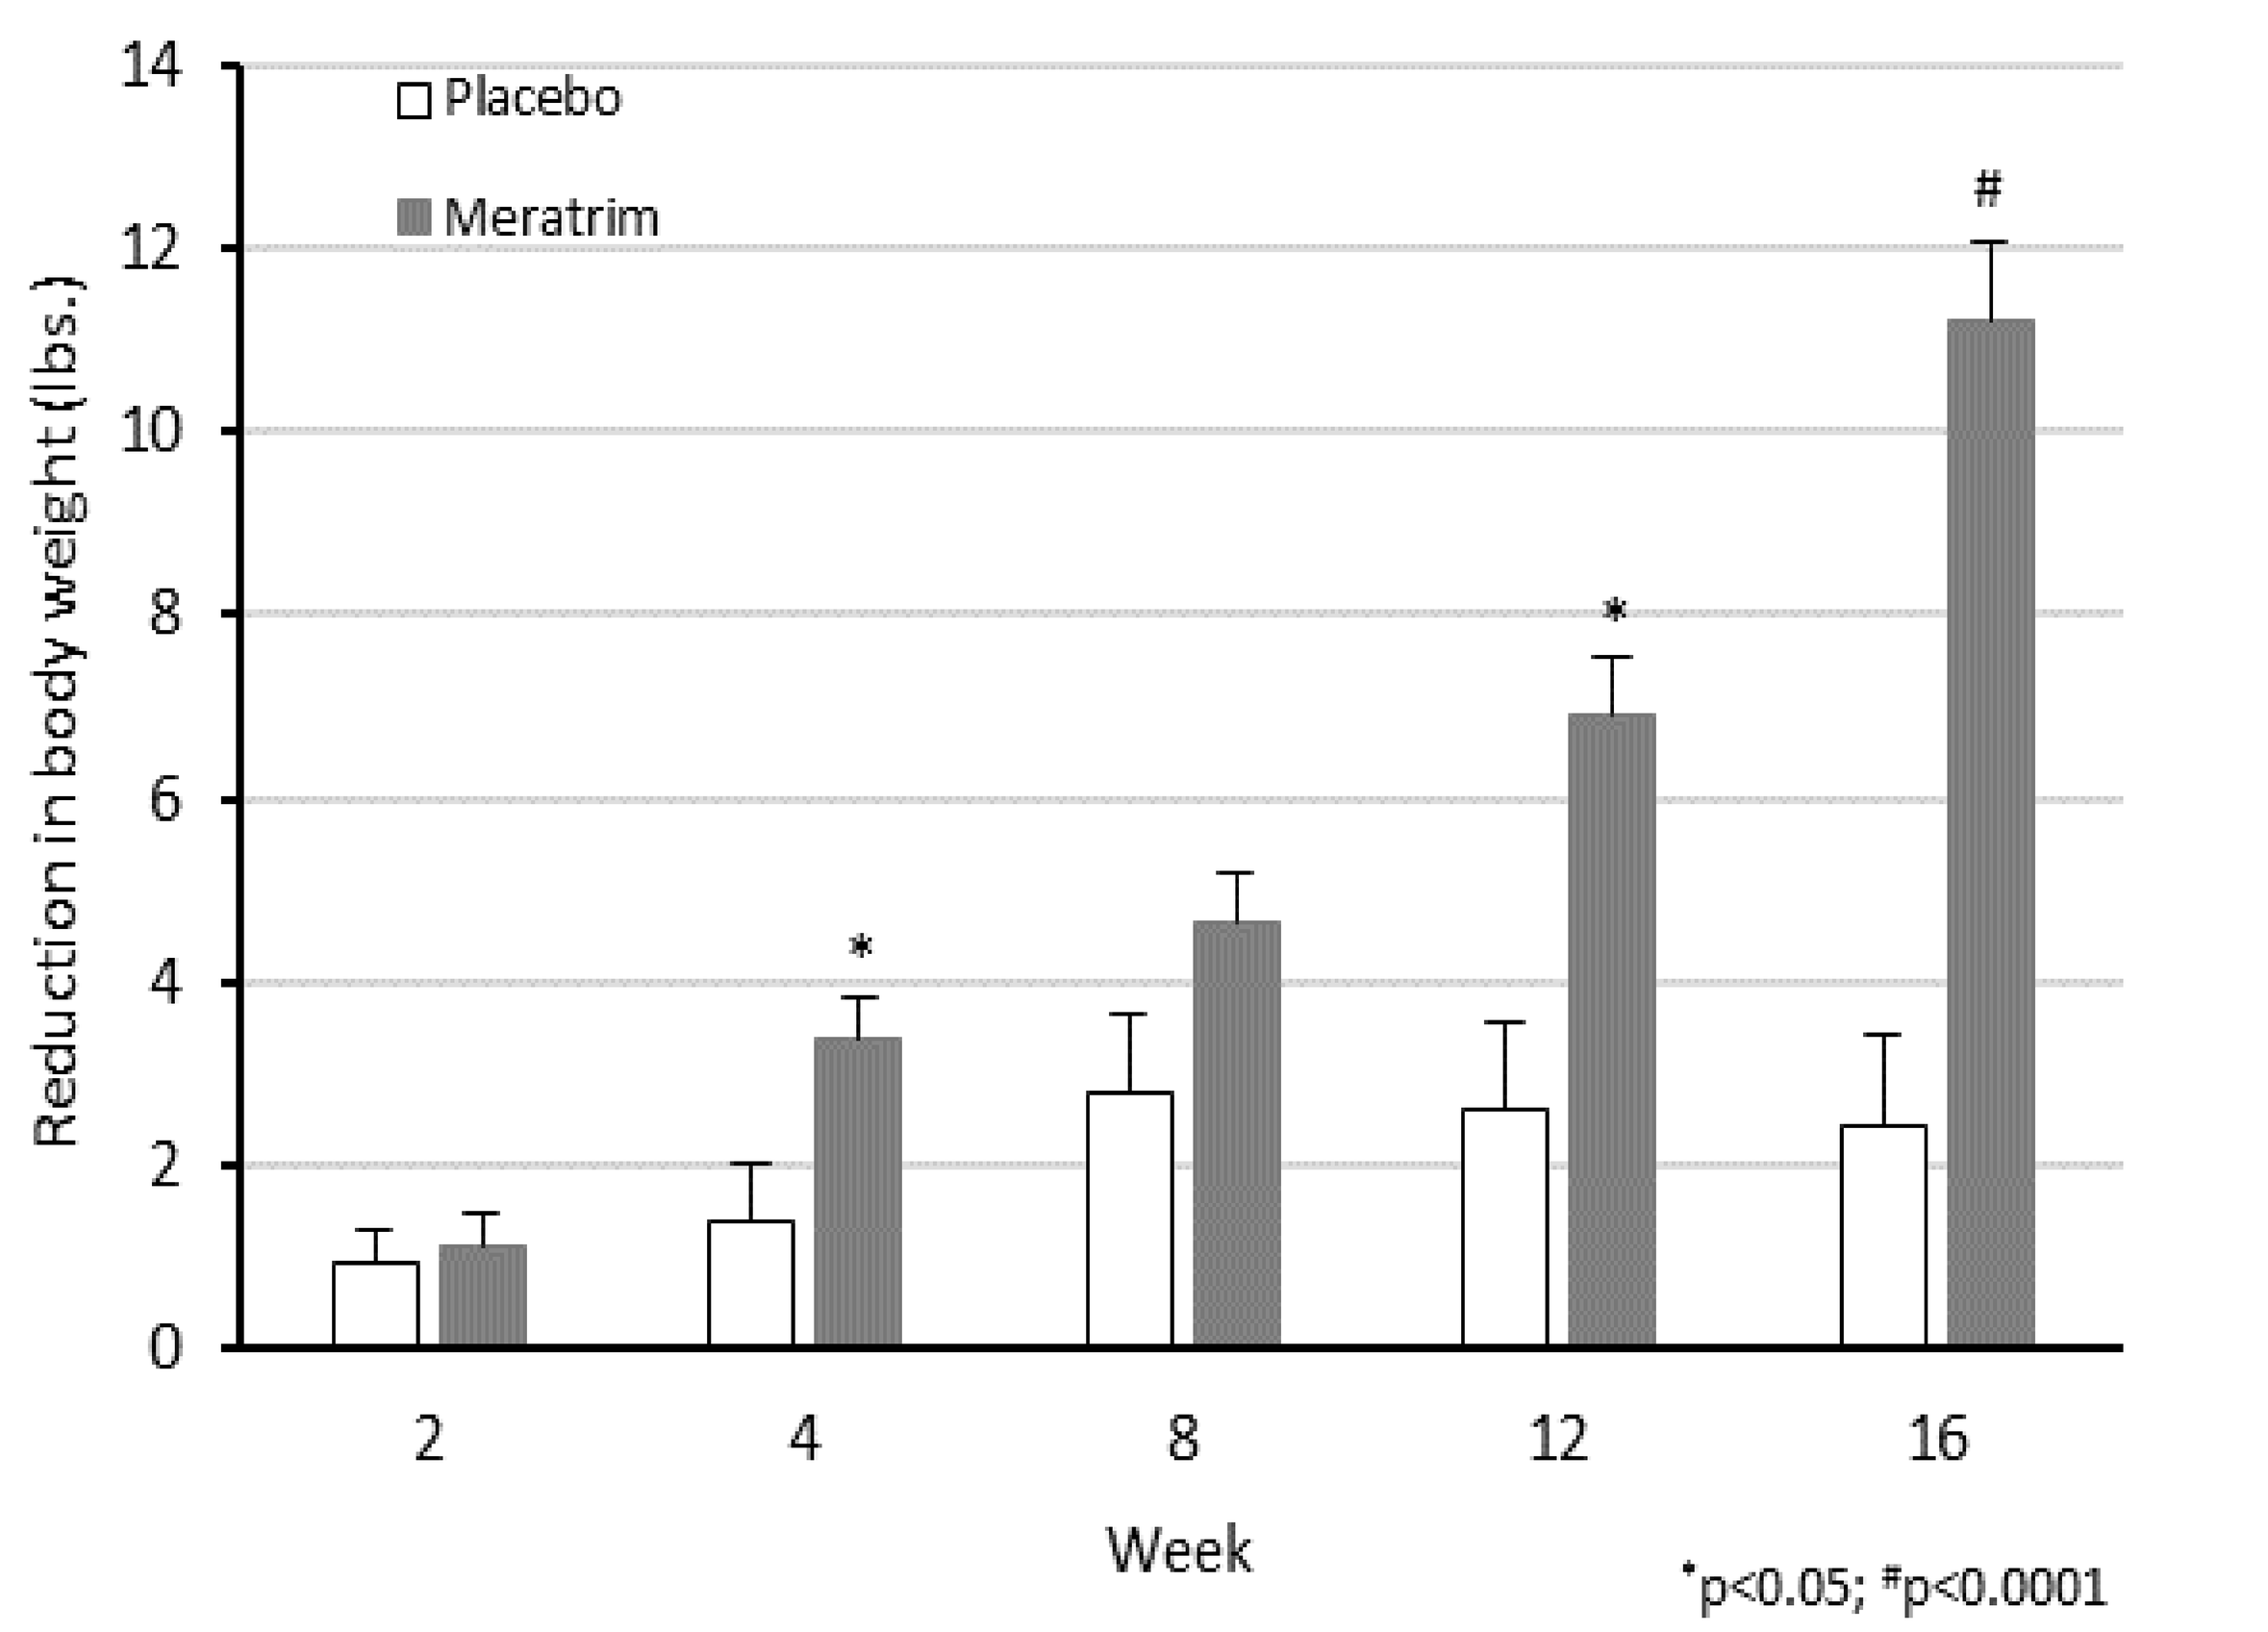
Placebo
Meratrim
Reduction in body weight (lbs.)
0
2
4
6
8
10
12
14
2
4
8
12
16
Week
*
*
#
*p<0.05; #p<0.0001

Figure 2: Reduction in waist & hip size (16 weeks)

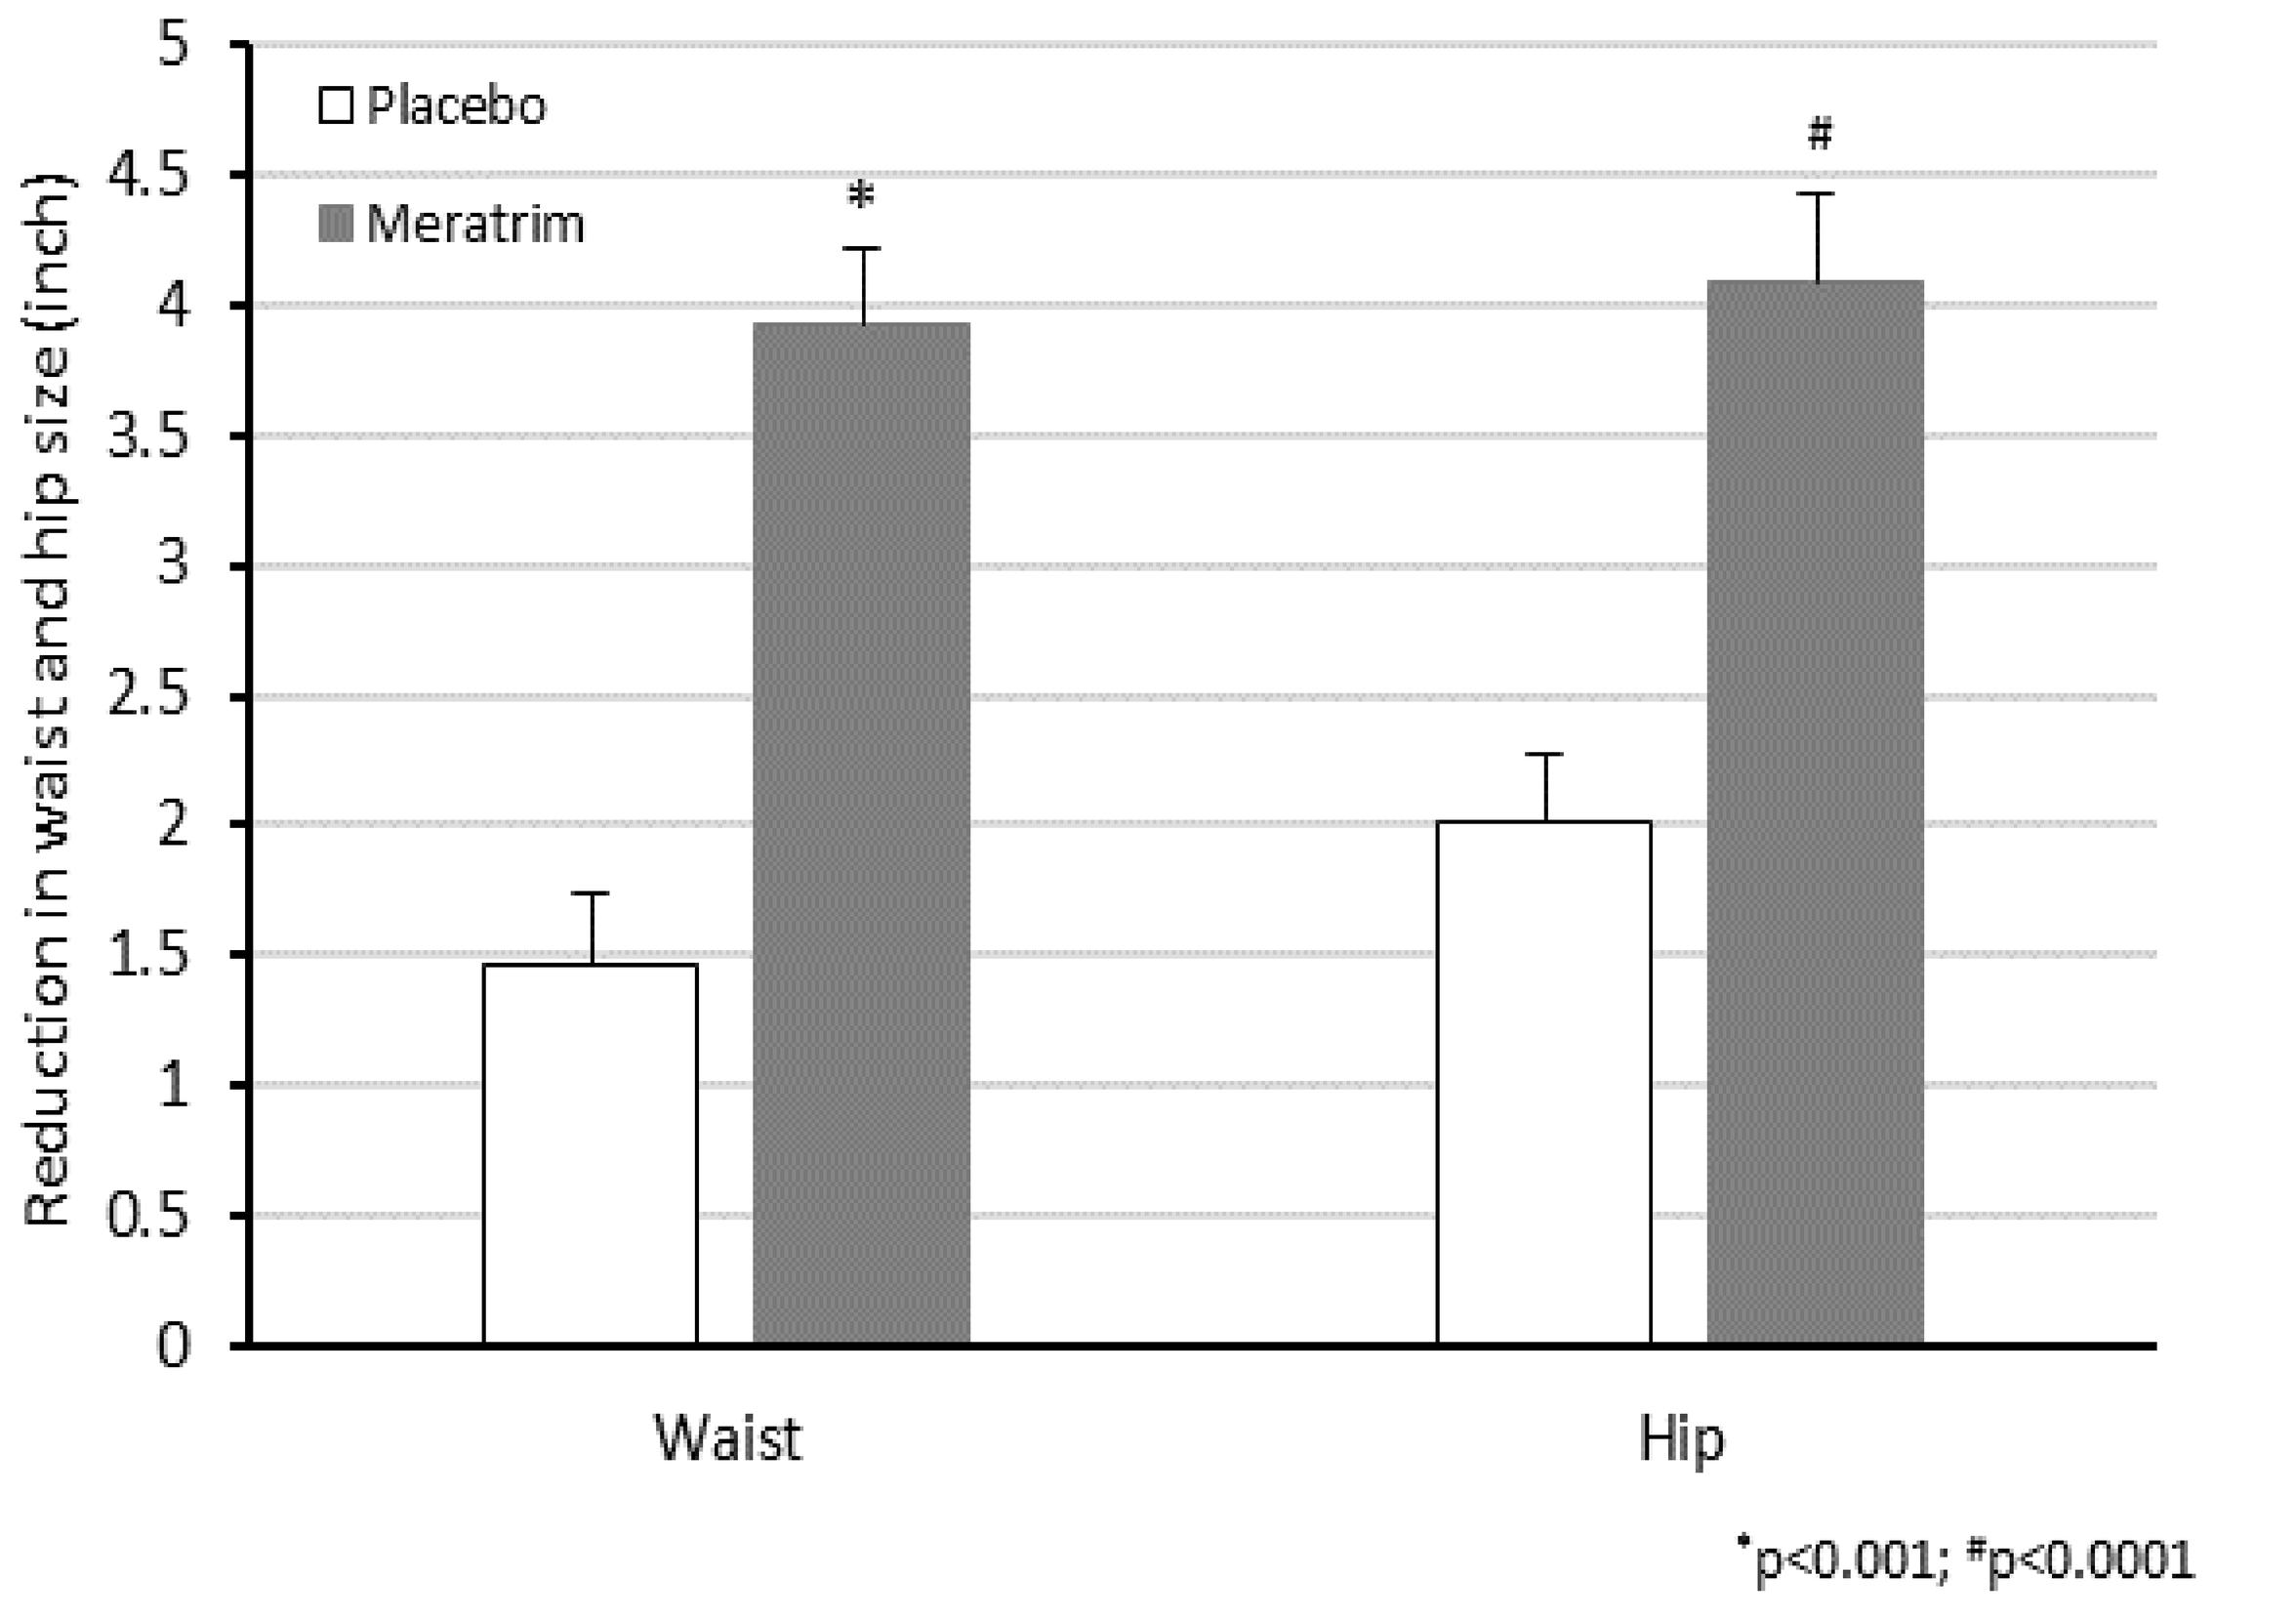
Placebo
Meratrim
Reduction in waist and hip size (inch)
0
0.5
1
1.5
2
2.5
3
3.5
4
4.5
5
Waist
Hip
*
#
*p<0.001; #p<0.0001

Table 1: Change in serum lipid profile after 16 weeks of supplementation

Parameter	Placebo (n=28)	Meratrim (n=29)	P value*
Triglycerides (mg/dL)	-13.68 ± 5.17	-43.62 ± 6.74	<0.001
Total Cholesterol (mg/dL)	-0.75 ± 5.10	-20.00 ± 2.88	0.0002
LDL (mg/dL)	6.25 ± 4.79	-14.79 ± 2.93	<0.0001
HDL (mg/dL)	-1.96 ± 0.48	2.03 ± 0.75	<0.0001
LDL-HDL Ratio	0.35 ± 0.14	-0.49 ± 0.11	<0.0001

*Significant difference between the Meratrim and the placebo group

CONCLUSIONS

- Meratrim appears to be well-tolerated and effective ingredient for weight management in healthy overweight subjects.

Presented at the 56th Annual Meeting of the American College of Nutrition, Nov 11-13, 2015, Orlando, FL